Selective peroxisome proliferator-activated receptor (PPAR) delta (δ) agonist, mavodelpar, improves cellular bioenergetics in fibroblasts from patients with mitochondrial complex I deficiency.

Bianca Seminotti[1*], John Adams[2*], Jerry Vockley[1,3,4*]

[1] Division of Genetic and Genomic Medicine, Department of Pediatrics, University of Pittsburgh School of Medicine, Pittsburgh, PA 15224, USA;
[2] Reneo Pharmaceuticals, Inc., 18575 Jamboree Road Suite 275-S, Irvine, CA 92612, USA; [3] Department of Human Genetics, University of Pittsburgh Graduate School of Public Health, Pittsburgh, PA 15261, USA; [4] UPMC Children's Hospital of Pittsburgh, Pittsburgh, PA 15224, USA.

***Corresponding email: seminotti.bianca@pitt.edu, jadams@reneopharma.com, vockleyg@upmc.edu**

OBJECTIVE

To evaluate whether Mavodelpar could either directly or indirectly improve energy metabolism in cells from CI-deficient patients with evidence of myopathy by decreasing reactive species production and improving OXPHOS efficiency.

MATERIALS & METHODS

The effect of mavodelpar was evaluated on superoxide generation and mitochondrial respiration in fibroblasts from CI-deficient patients with mutations in the genes *ACAD9*, *NDUFV1* (nuclear DNA) and *ND6* (mitochondrial DNA). Fibroblasts from healthy adults served as controls. Cells were cultured in medium without glucose for 48 hours to assess their ability to accommodate a shift of energy source from glucose to fatty acids. The effect of mavodelpar on superoxide production and oxygen consumption rate (OCR) was measured with MitoSox Red dye and a Seahorse Bioanalyzer, respectively.

BACKGROUND

Mitochondrial complex I (CI) deficiency is the most frequent cause of oxidative phosphorylation (OXPHOS) disorders in humans. CI deficiency is clinically heterogeneous. Many patients develop musculoskeletal, neurologic, and cardiac symptoms during their early years of life and have a rapidly progressive disease course, often resulting in fatal outcomes [1,2,3]. Adult onset of CI deficiency is also reported [1,2,3]. Clinical symptoms include hypotonia, myopathy, seizures, cardiomyopathy, Leigh and Leigh-like syndromes, life threatening infantile lactic acidosis, leukodystrophic encephalopathy, and developmental delay. Peroxisome proliferator-activated receptor (PPAR) deltas are nuclear receptors/transcription factors that modulate gene expression, regulating mitochondrial energy and metabolic homeostasis. Mavodelpar is a selective PPARδ agonist currently in clinical development for PMM treatment.

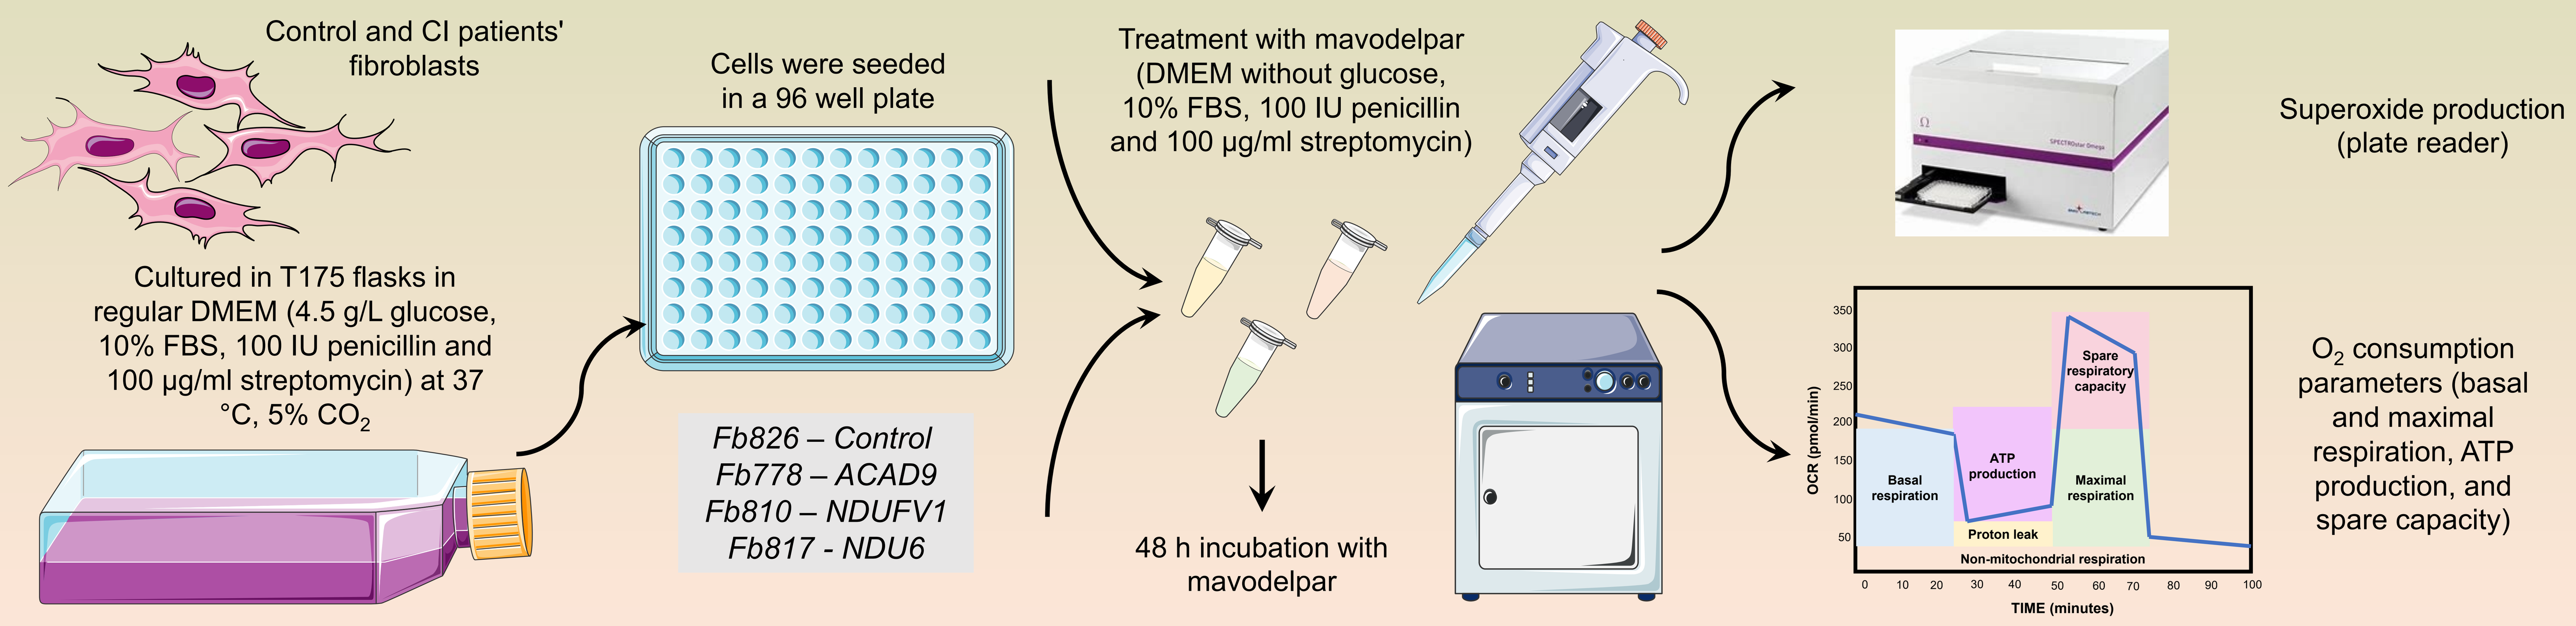
Control and CI patients' fibroblasts
Cultured in T175 flasks in regular DMEM (4.5 g/L glucose, 10% FBS, 100 IU penicillin and 100 µg/ml streptomycin) at 37 °C, 5% CO_2
Cells were seeded in a 96 well plate
Fb826 – Control
Fb778 – ACAD9
Fb810 – NDUFV1
Fb817 - NDU6
Treatment with mavodelpar (DMEM without glucose, 10% FBS, 100 IU penicillin and 100 µg/ml streptomycin)
48 h incubation with mavodelpar
Superoxide production (plate reader)
O_2 consumption parameters (basal and maximal respiration, ATP production, and spare capacity)
OCR (pmol/min)
Basal respiration
ATP production
Spare respiratory capacity
Maximal respiration
Proton leak
Non-mitochondrial respiration
TIME (minutes)

RESULTS

In untreated cells, superoxide generation was increased in CI-deficient patient fibroblasts compared to healthy fibroblast controls (Figure 1), whereas OCR and adenosine triphosphate (ATP) levels were decreased (Figure 2). Improved cellular bioenergetics were observed in all mavodelpar-treated CI-deficient cells including decreased superoxide levels (Figure 1B, C and D), and increased OCR and ATP production (Figures 3, 4, 5 and 6).

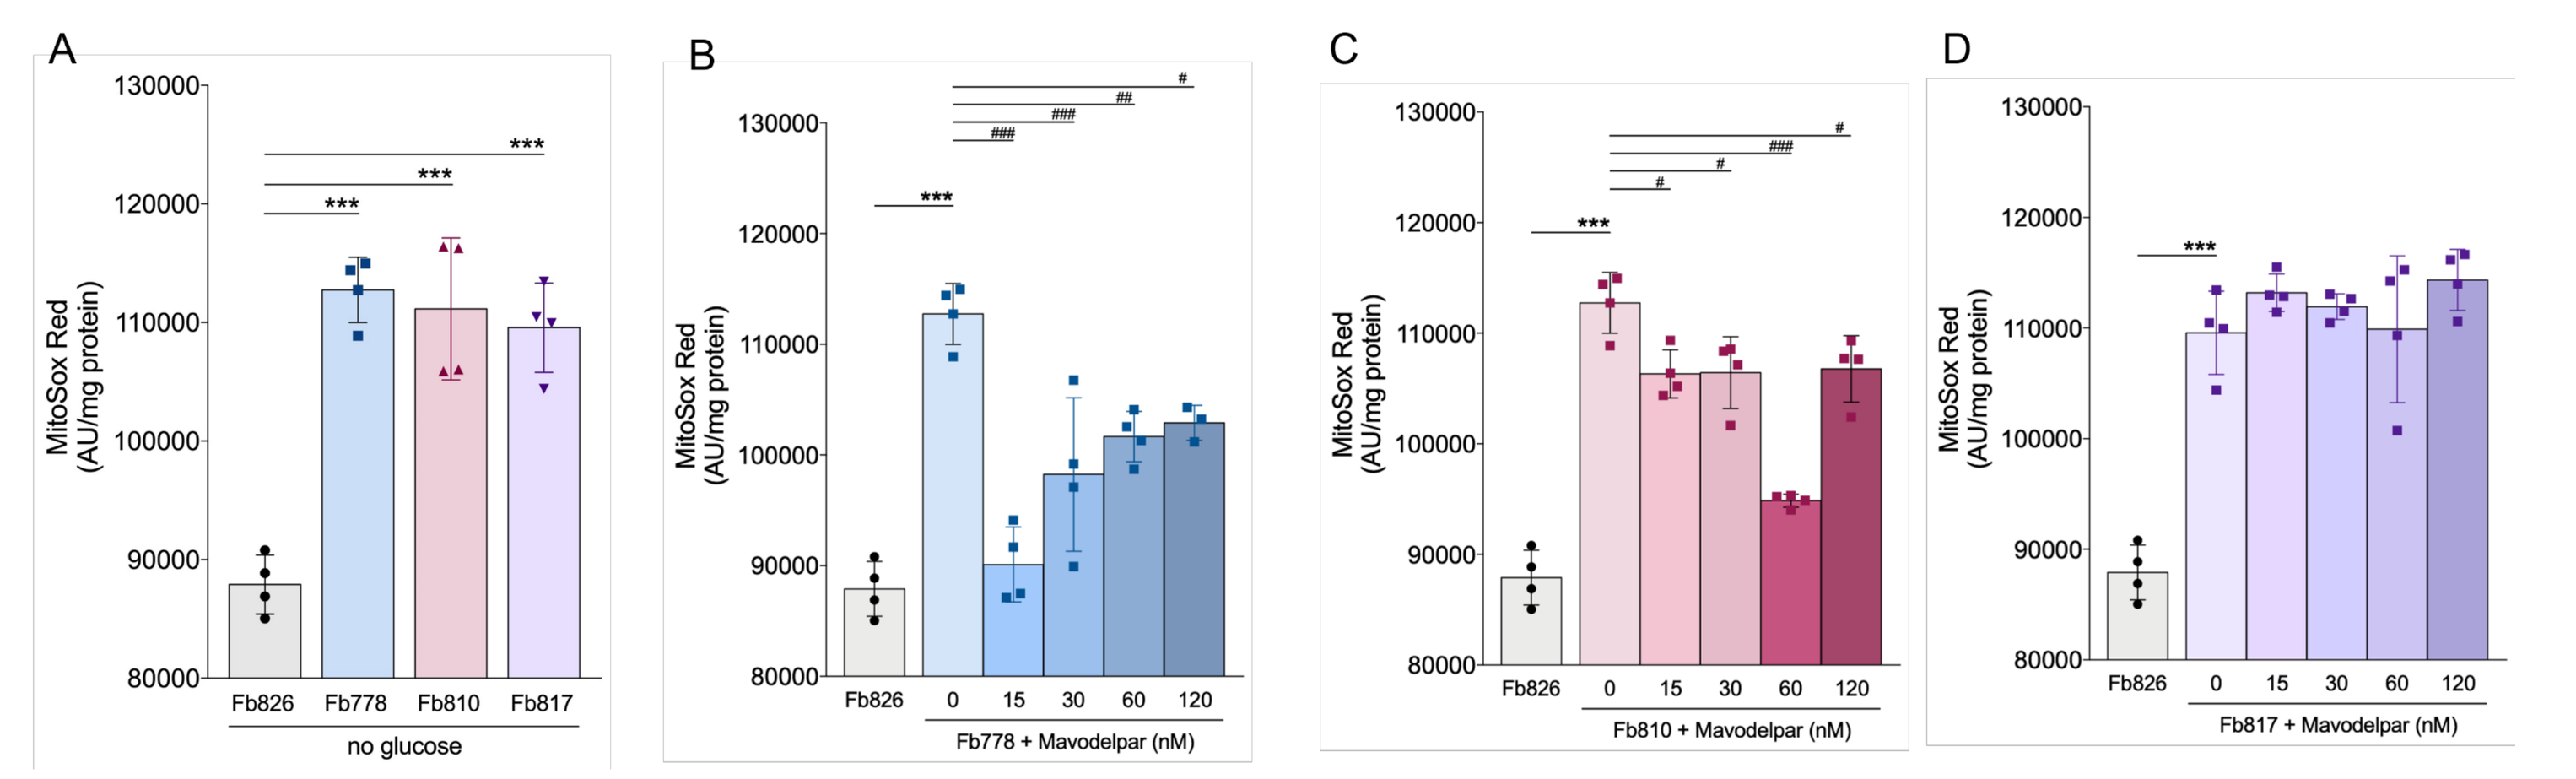
A
B
C
D
MitoSox Red (AU/mg protein)
Fb826
Fb778
Fb810
Fb817
no glucose
Fb778 + Mavodelpar (nM)
Fb810 + Mavodelpar (nM)
Fb817 + Mavodelpar (nM)

Figure 1. Superoxide production in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) in response to 48 hours mavodelpar treatment in the absence of glucose. Values were calculated after normalizing AU values to mg protein. Data are means ± SD; number of replicates: 6–8. **P* < 0.05, ***P* < 0.01, ****P* < 0.001 compared to control cells (Fb826); #P < 0.05, ##P < 0.01, ###P < 0.001, compared to untreated cells (One-way ANOVA followed by Tukey multiple range test).

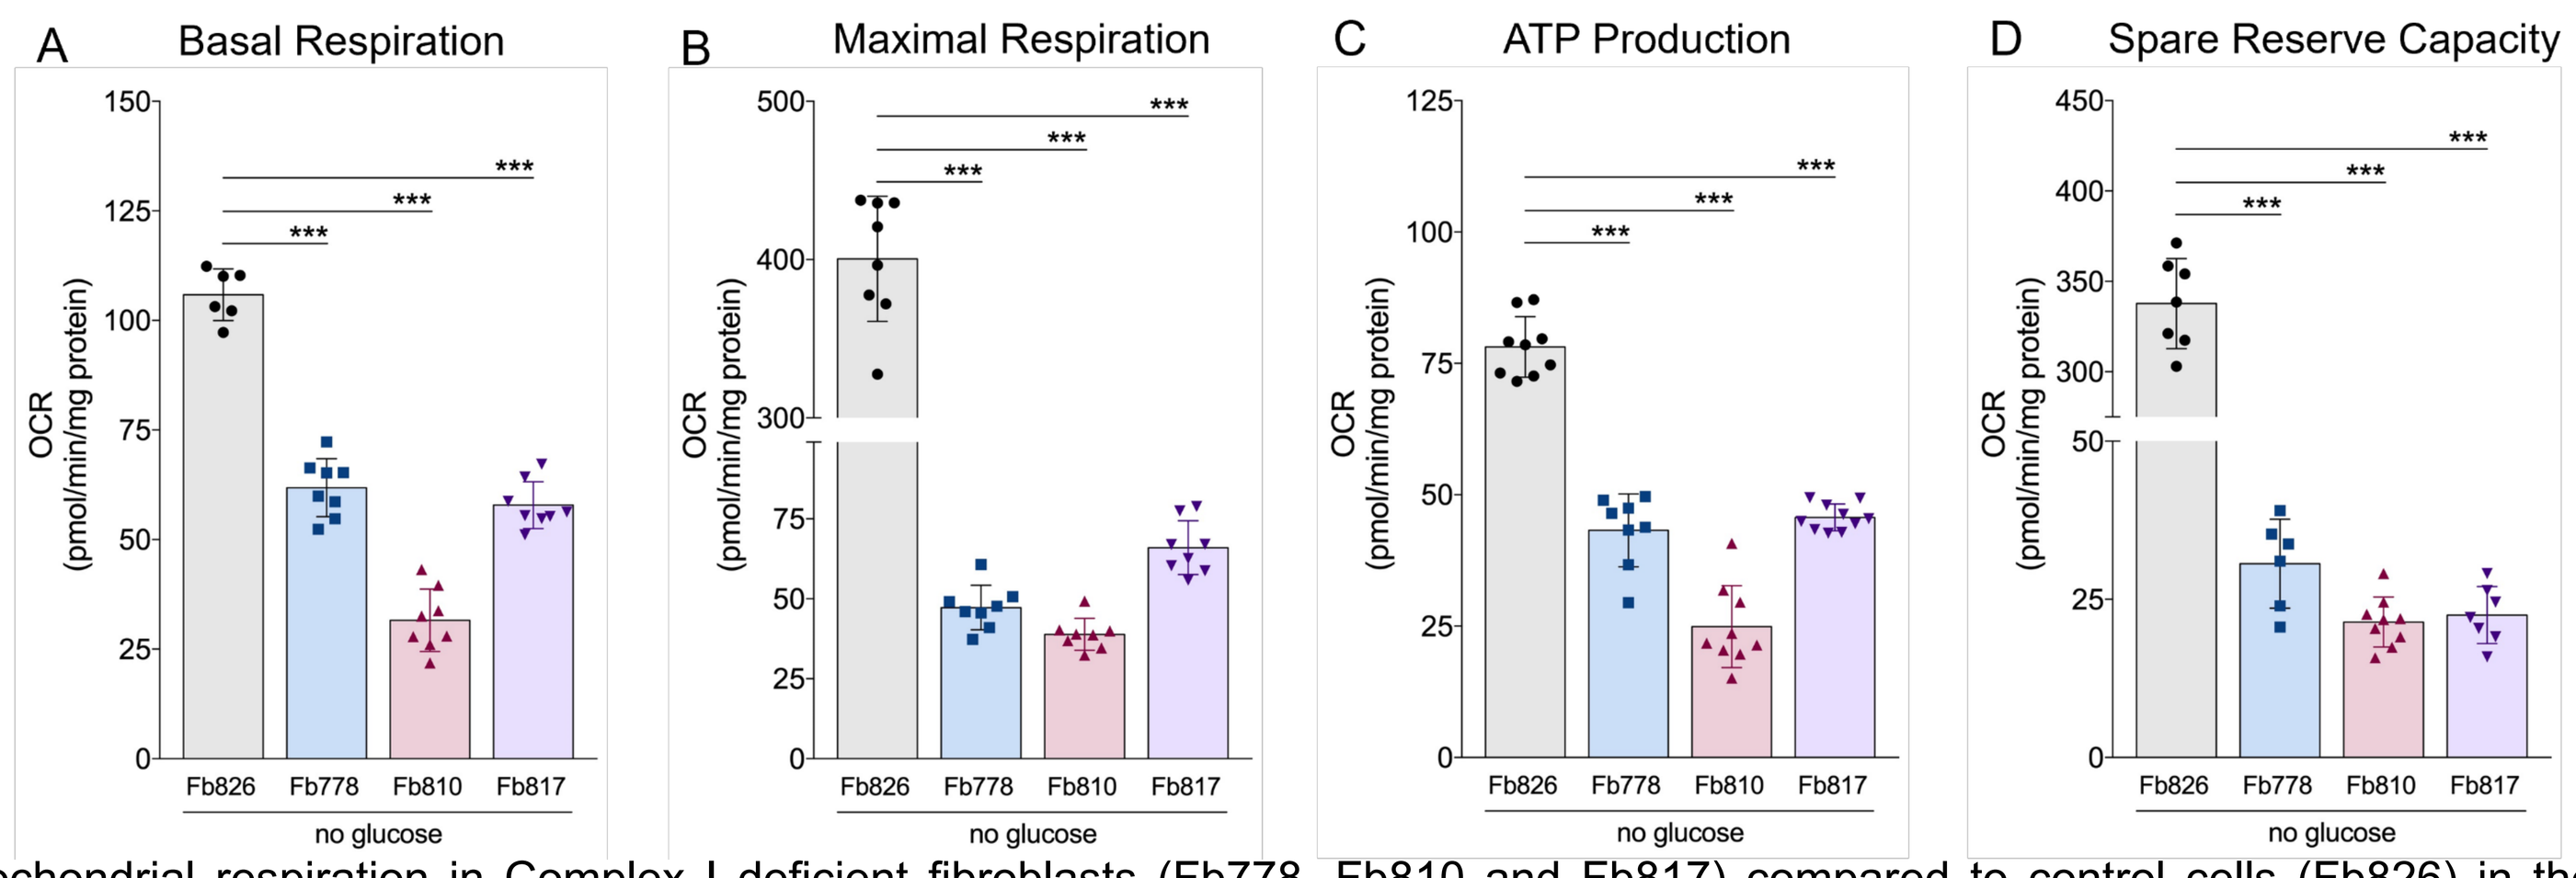
A Basal Respiration
B Maximal Respiration
C ATP Production
D Spare Reserve Capacity
OCR (pmol/min/mg protein)
Fb826
Fb778
Fb810
Fb817
no glucose

Figure 2. Mitochondrial respiration in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) compared to control cells (Fb826) in the absence of glucose. Oxygen consumption rate (OCR) was measured in resting state followed by sequential injection of Oligomycin, FCCP, and Rotenone/Antimycin A. The key assay parameter values were calculated after normalizing OCR values to mg protein. Basal respiration, Maximal Respiration, ATP production and Spare Reserve Capacity. Data are means ± SD; number of replicates: 6–8. **P* < 0.05, ***P* < 0.01, ****P* < 0.001 (One-way ANOVA followed by Tukey multiple range test).

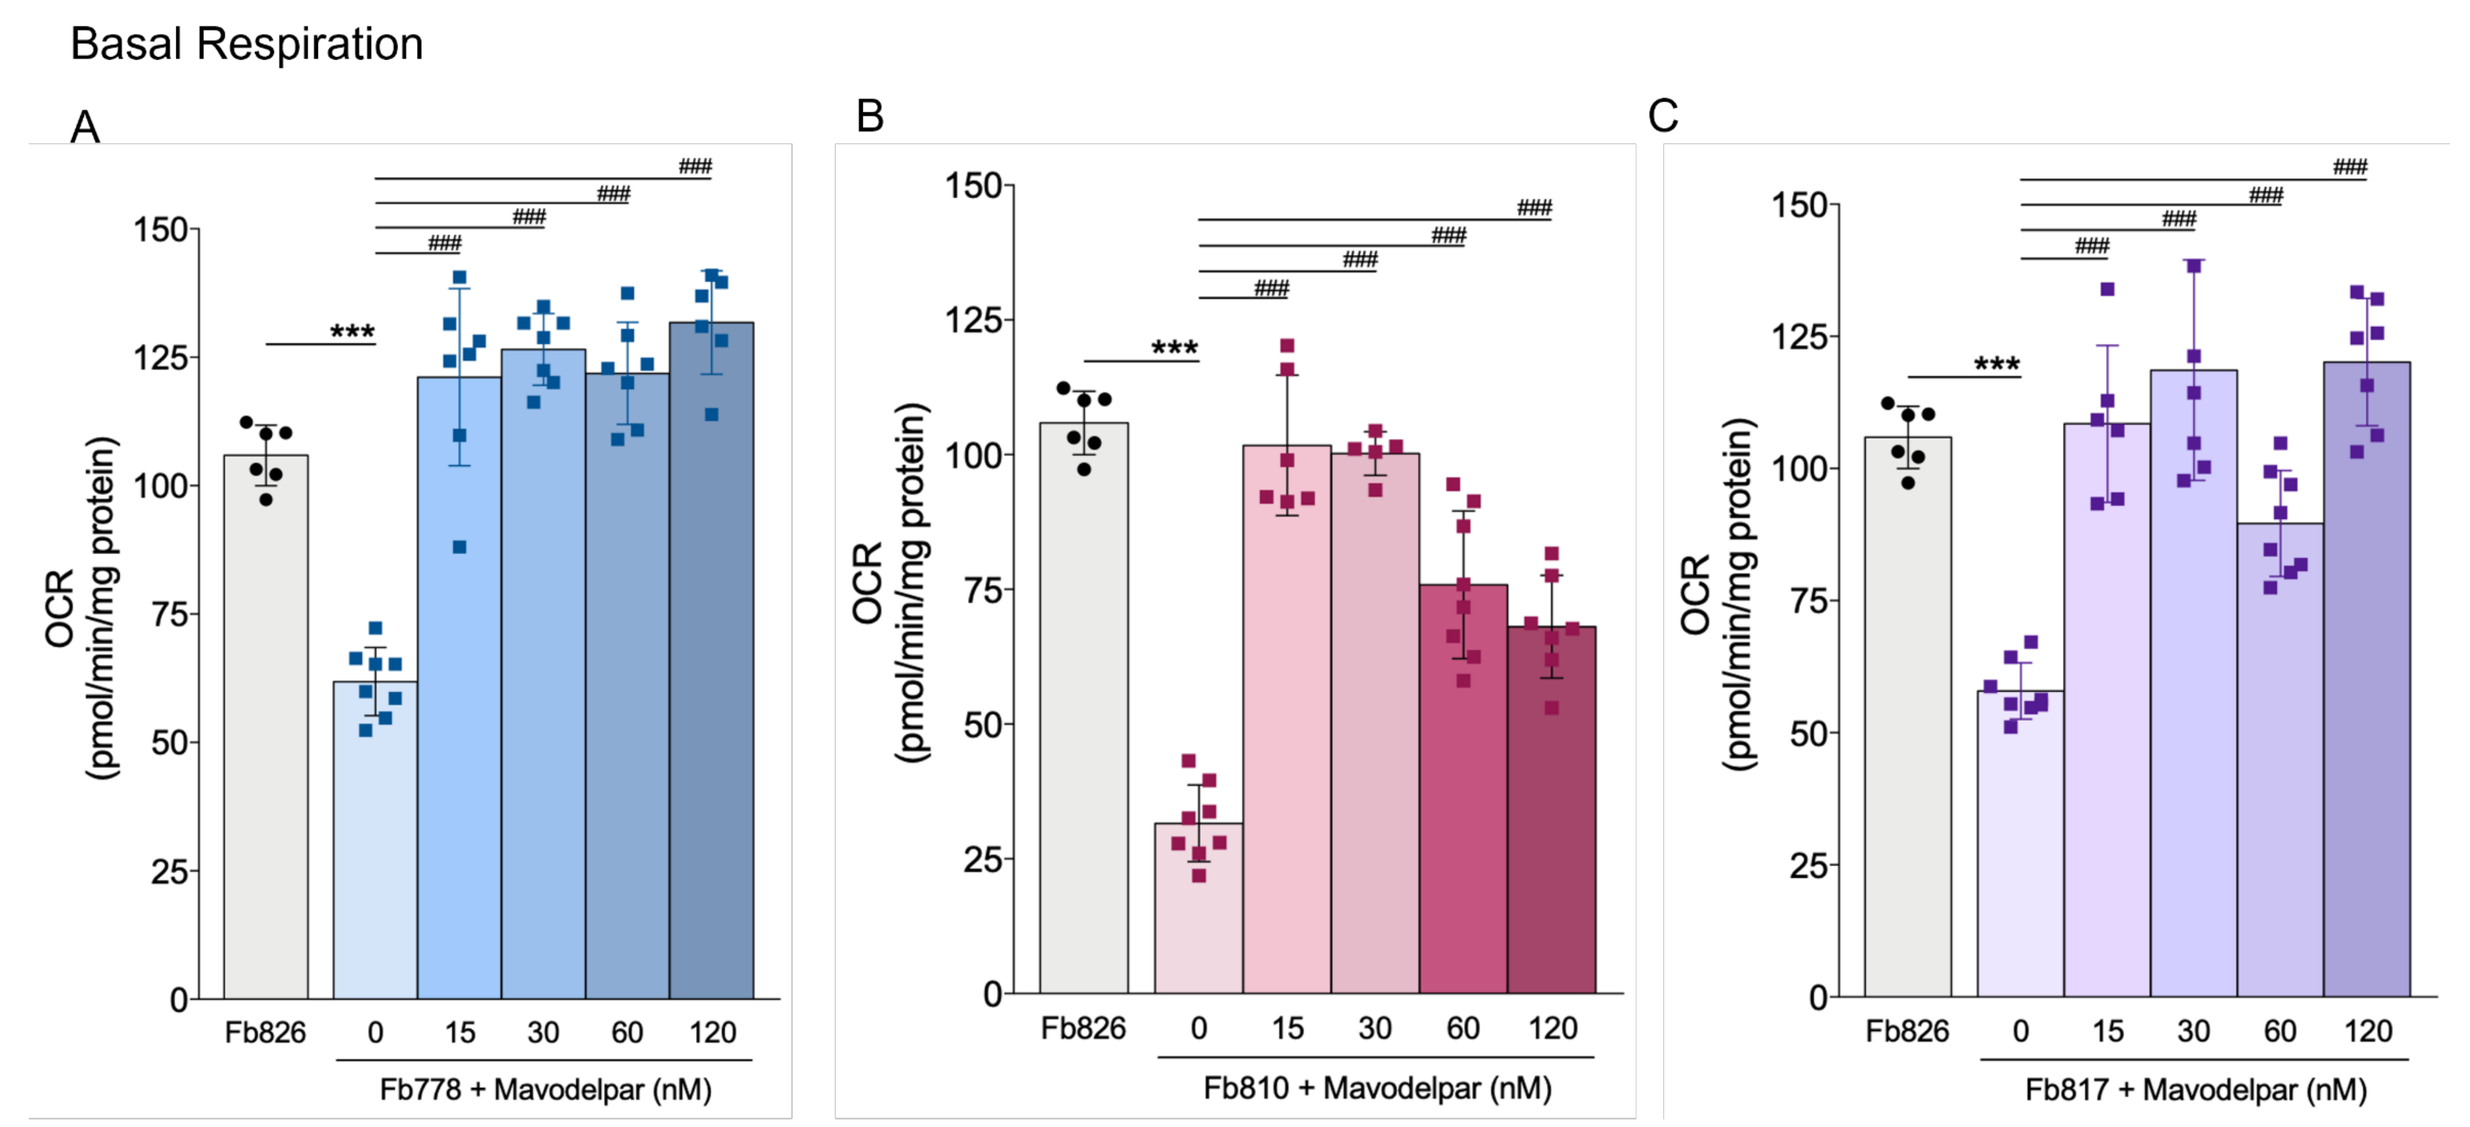
Basal Respiration
A
B
C
OCR (pmol/min/mg protein)
Fb826
Fb778 + Mavodelpar (nM)
Fb810 + Mavodelpar (nM)
Fb817 + Mavodelpar (nM)

Figure 3. Mitochondrial respiration in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) in response to 48 hours mavodelpar treatment in the absence of glucose. Oxygen consumption rate (OCR) for basal respiration was measured in resting state. Values were calculated after normalizing OCR values to mg protein. Data are means ± SD; number of replicates: 6–8. *P < 0.05, **P < 0.01, ***P < 0.001 compared to control cells (Fb826); #P < 0.05, ##P < 0.01, ###P < 0.001, compared to untreated cells (One-way ANOVA followed by Tukey multiple range test).

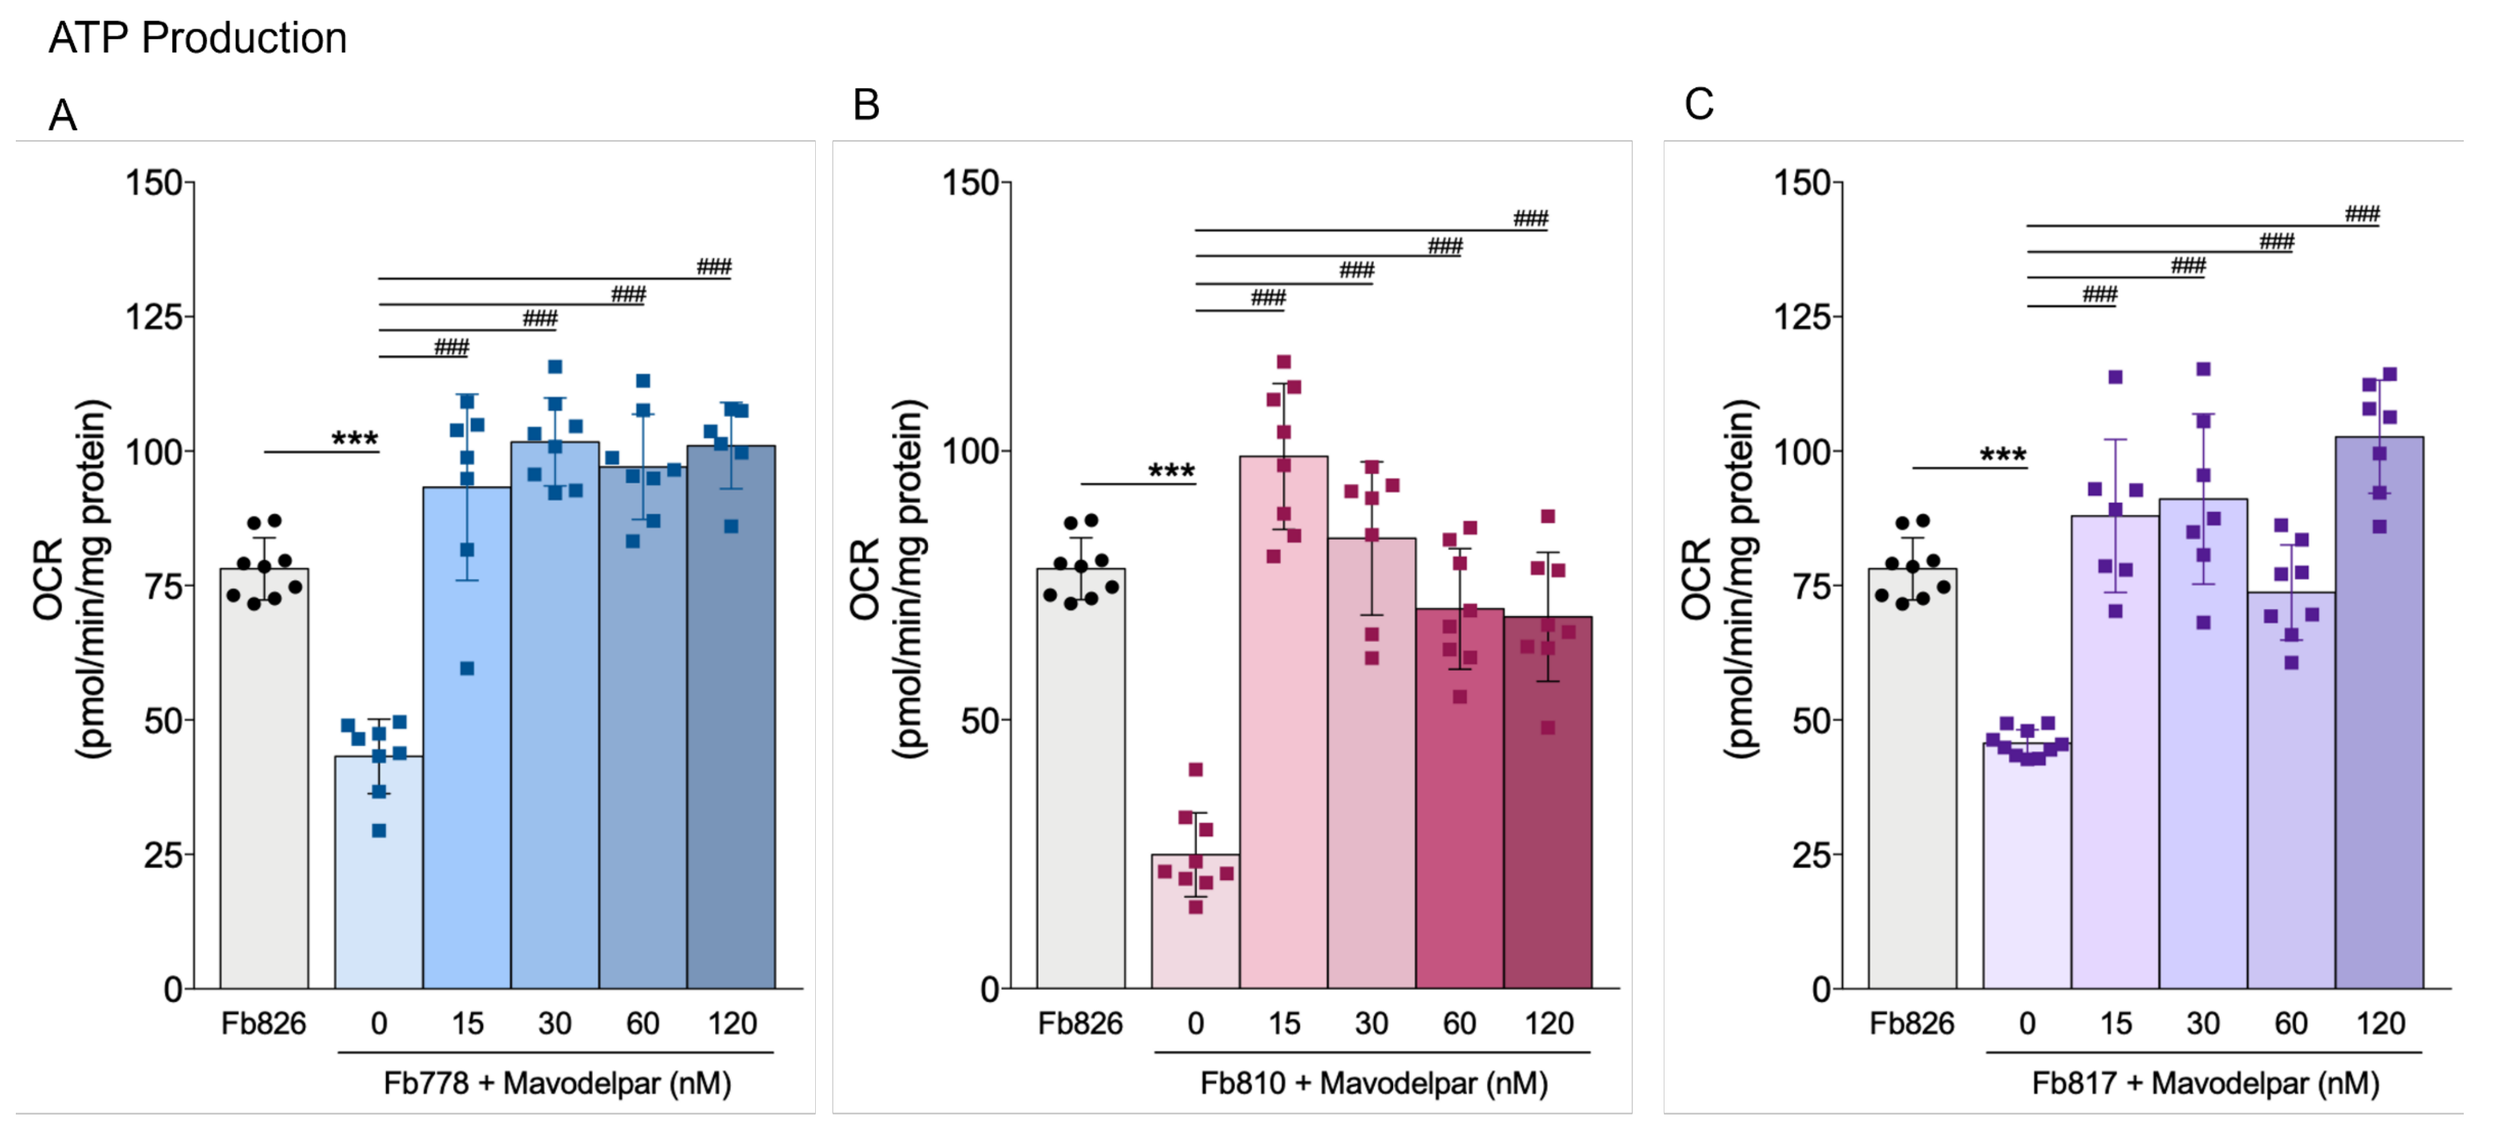
ATP Production
A
B
C
OCR (pmol/min/mg protein)
Fb826
Fb778 + Mavodelpar (nM)
Fb810 + Mavodelpar (nM)
Fb817 + Mavodelpar (nM)

Figure 5. Mitochondrial respiration in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) in response to 48 hours mavodelpar treatment in the absence of glucose. Oxygen consumption rate (OCR) for ATP production was measured after Oligomycin injection. Values were calculated after normalizing OCR values to mg protein. Data are means ± SD; number of replicates: 6–8. *P < 0.05, **P < 0.01, ***P < 0.001 compared to control cells (Fb826); #P < 0.05, ##P < 0.01, ###P < 0.001, compared to untreated cells (One-way ANOVA followed by Tukey multiple range test).

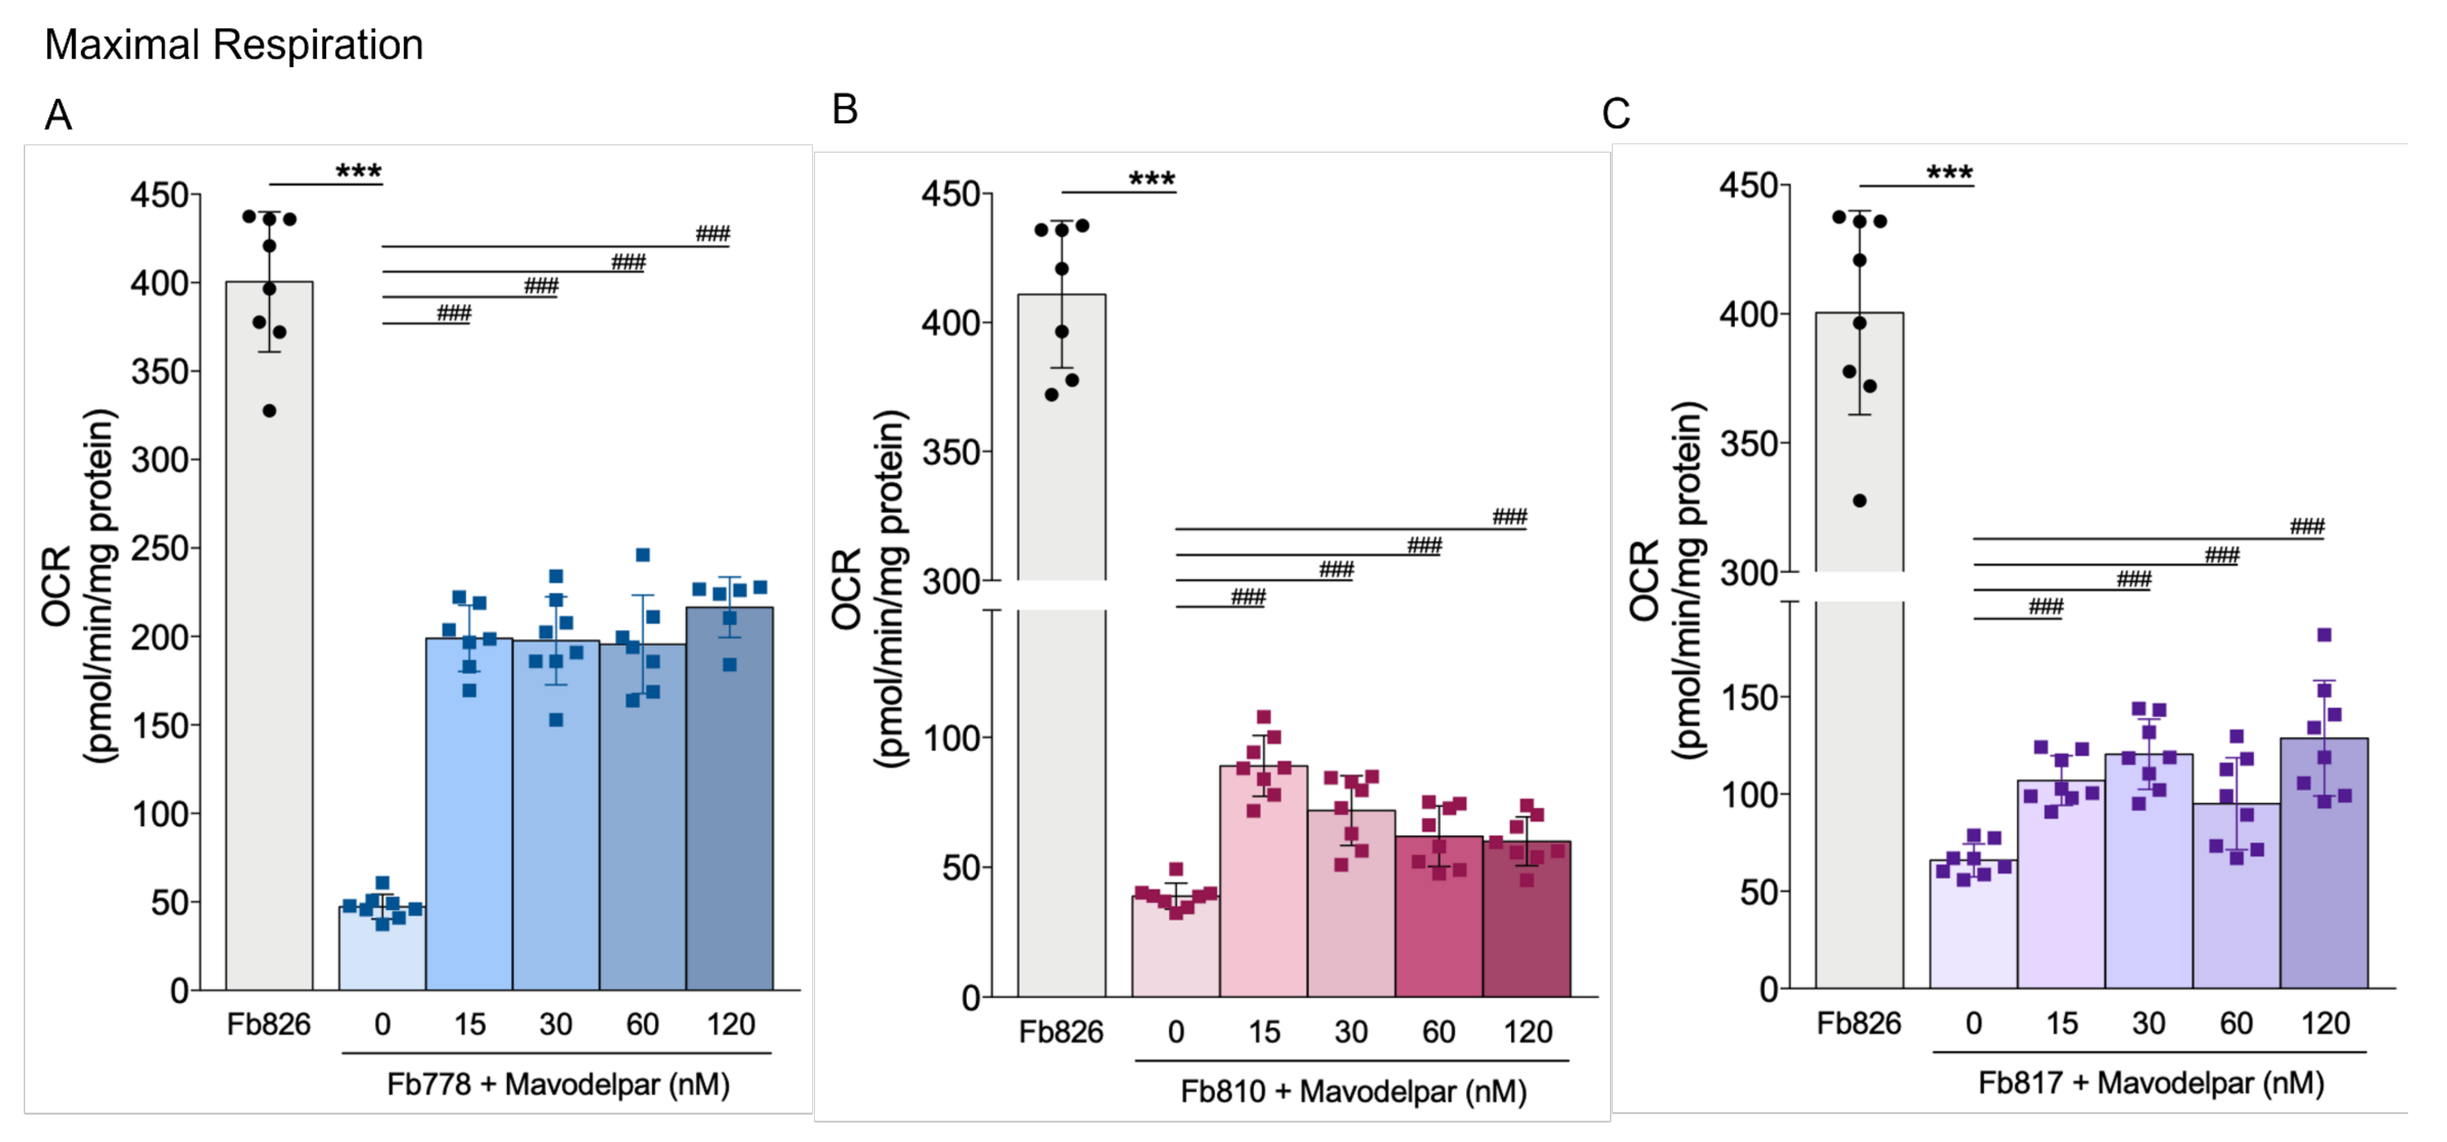
Maximal Respiration
A
B
C
OCR (pmol/min/mg protein)
Fb826
Fb778 + Mavodelpar (nM)
Fb810 + Mavodelpar (nM)
Fb817 + Mavodelpar (nM)

Figure 4. Mitochondrial respiration in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) in response to 48 hours mavodelpar treatment in the absence of glucose. Oxygen consumption rate (OCR) for maximal respiration was measured after FCCP injection. Values were calculated after normalizing OCR values to mg protein. Data are means ± SD; number of replicates: 6–8. *P < 0.05, **P < 0.01, ***P < 0.001 compared to control cells (Fb826); #P < 0.05, ##P < 0.01, ###P < 0.001, compared to untreated cells (One-way ANOVA followed by Tukey multiple range test).

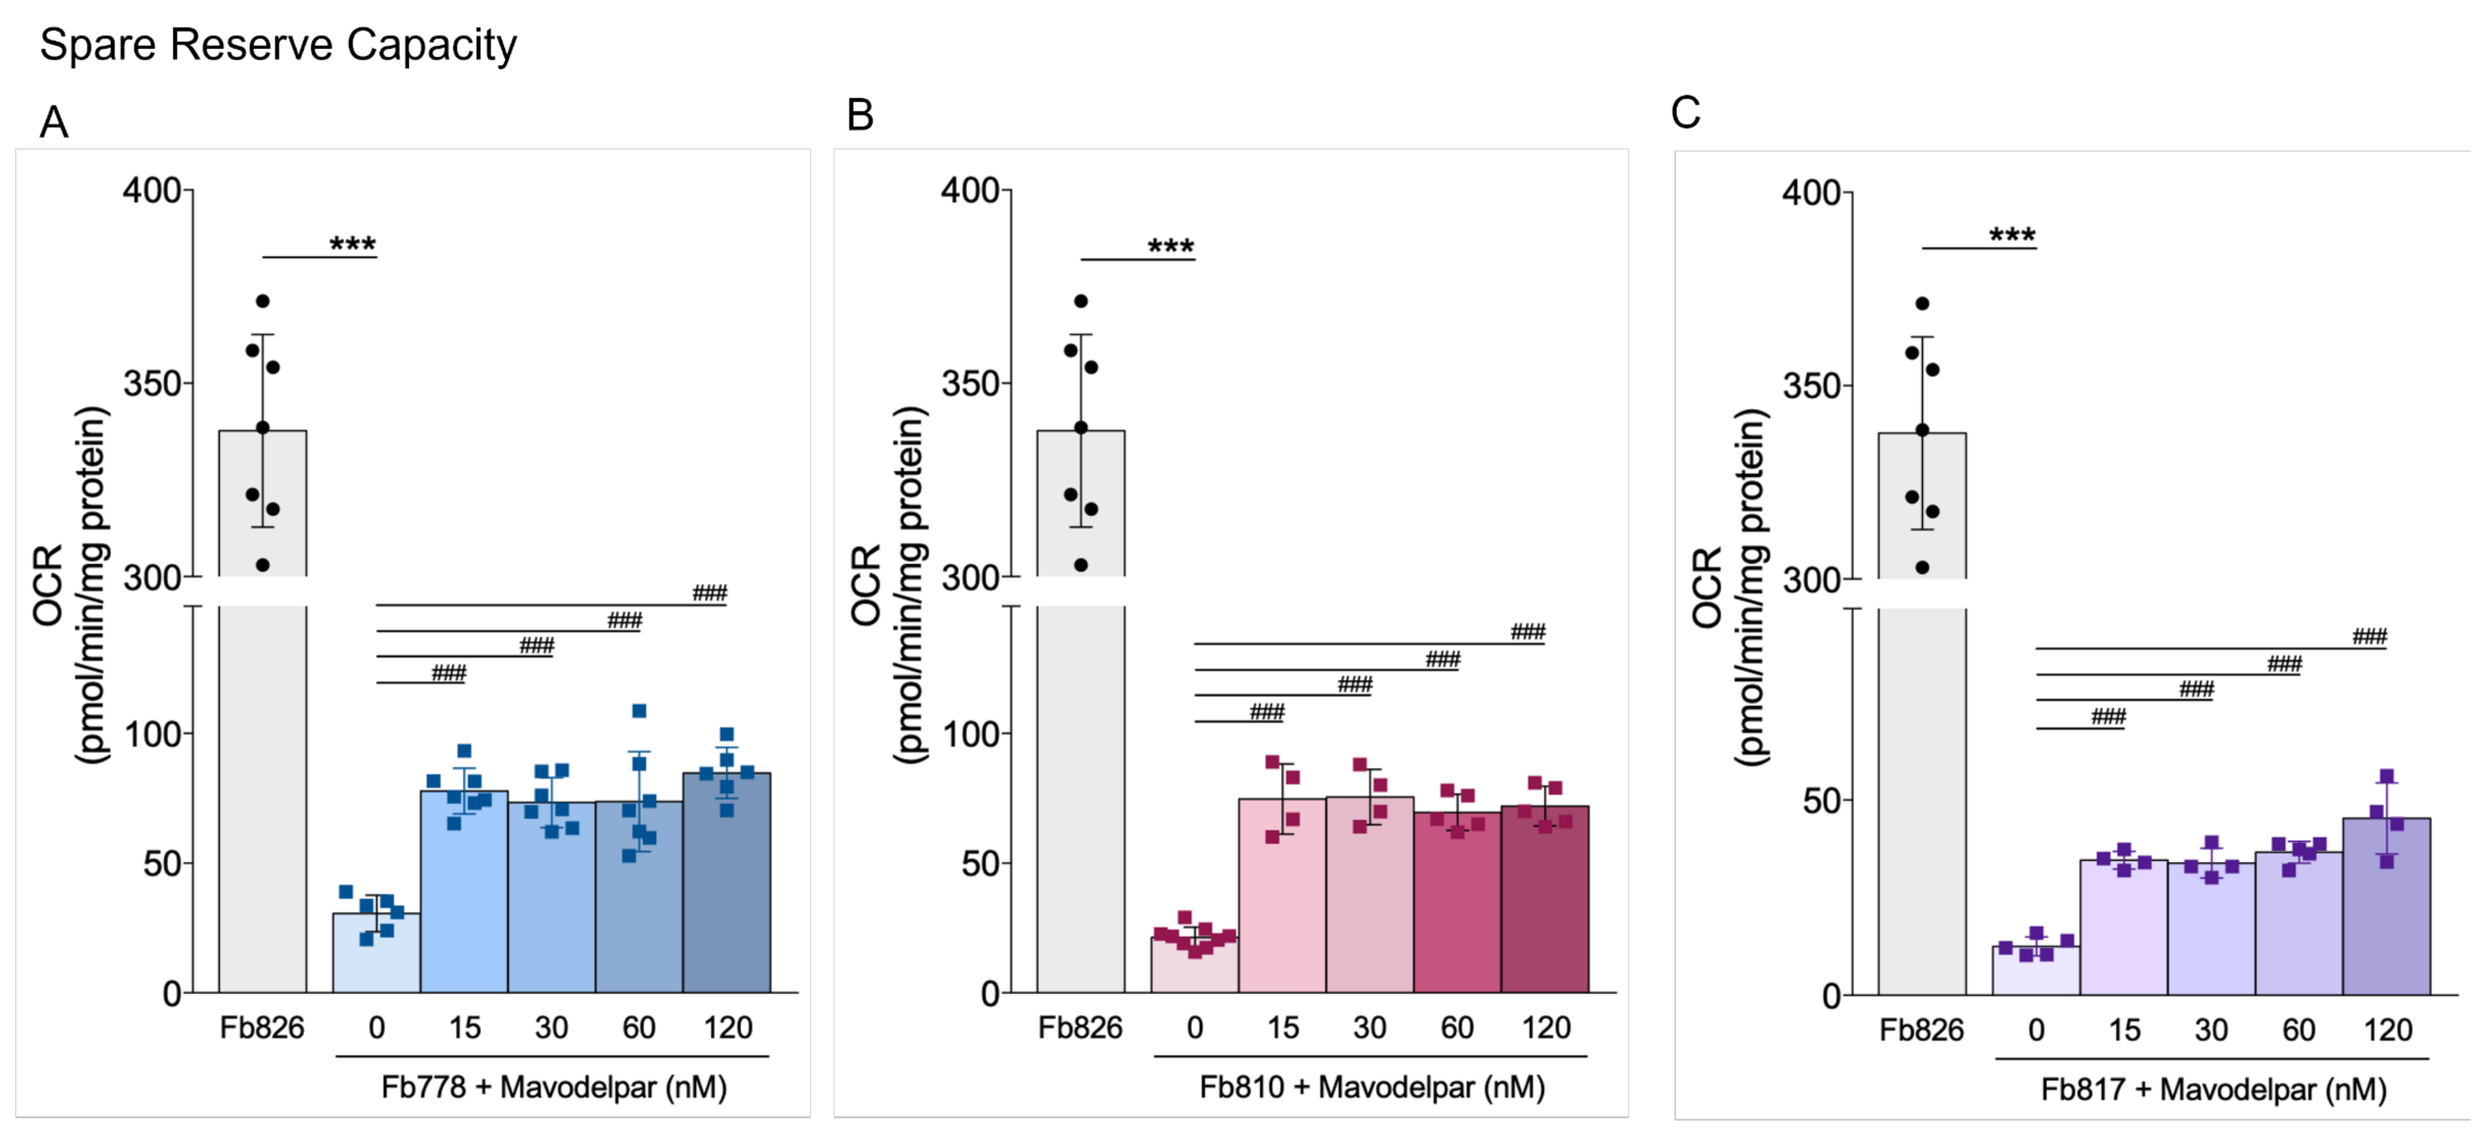
Spare Reserve Capacity
A
B
C
OCR (pmol/min/mg protein)
Fb826
Fb778 + Mavodelpar (nM)
Fb810 + Mavodelpar (nM)
Fb817 + Mavodelpar (nM)

Figure 6. Mitochondrial respiration in Complex I deficient fibroblasts (Fb778, Fb810 and Fb817) in response to 48 hours mavodelpar treatment in the absence of glucose. Oxygen consumption rate (OCR) for spare reserve capacity was obtained after subtracting basal respiration from maximal respiratory capacity. Values were calculated after normalizing OCR values to mg protein. Data are means ± SD; number of replicates: 6–8. *P < 0.05, **P < 0.01, ***P < 0.001 compared to control cells (Fb826); #P < 0.05, ##P < 0.01, ###P < 0.001, compared to untreated cells (One-way ANOVA followed by Tukey multiple range test).

DISCUSSION AND CONCLUSION

These findings demonstrate that the bioenergetic deficiencies and mitochondrial dysfunction observed in CI-deficient patient fibroblasts can potentially be alleviated by treatment with mavodelpar. Further, because mavodelpar activation results in increased transcription of genes involved in mitochondrial biogenesis, fatty acid oxidation and energy production in the form of ATP [4], these results provide mechanistic evidence for the potential of mavodelpar as a therapeutic option in patients with mitochondrial complex I deficiency and Primary Mitochondrial Myopathies (PMM).

REFERENCES

[1] Koene, S et al. "Natural disease course and genotype-phenotype correlations in Complex I deficiency caused by nuclear gene defects: what we learned from 130 cases." Journal of inherited metabolic disease vol. 35,5 (2012): 737-47. doi:10.1007/s10545-012-949;. [2] Fassone, Elisa, and Shamima Rahman. "Complex I deficiency: clinical features, biochemistry and molecular genetics." Journal of medical genetics vol. 49,9 (2012): 578-90. doi:10.1136/jmedgenet-2012-101159; [3] Aintablian, H K et al. "An atypical presentation of ACAD9 deficiency: Diagnosis by whole exome sequencing broadens the phenotypic spectrum and alters treatment approach." Molecular genetics and metabolism reports vol. 10 38-44. 29 Dec. 2016, doi:10.1016/j.ymgmr.2016.12.005; [4] D'Annibale et al. Treatment of VLCAD-Deficient Patient Fibroblasts with Peroxisome Proliferator-Activated Receptor δ Agonist Improves Cellular Bioenergetics. Cells. 2022 Aug 24;11(17):2635. doi: 10.3390/cells11172635.